Effects of Hexametaphosphate Dentifrice on Stain Removal and Calculus

K.M. Kozak, D.J. White, S. Farrell*

P&G, Mason, OH, USA

0692

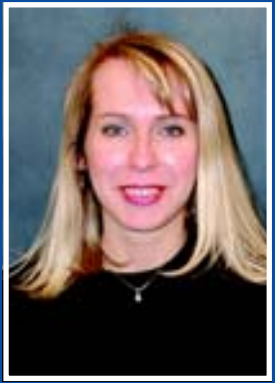

ABSTRACT

Hexametaphosphate dentifrices have been clinically proven to provide efficacy in tooth whitening (extrinsic stain removal and prevention) as well as in prevention accumulation of calcified tartar deposits. HMP is a 'calcium phosphate surface active builder' – meaning its solution calcium binding efficiencies are complemented by surface reactivity with minerals permitting intraoral retention. **Objectives:** In these studies the retentive actions of HMP on extrinsic stain removal and on prevention or calculus deposition were evaluated in specialized protocols *in vitro*. **Methods:** Bovine enamel blocks stained via the Stookey PCR method were pre-measured for CIELAB L* color and then cycled through a treatment sequence where they were chemically treated with water dentifrice supernates (treatments including Crest Cavity Protection control dentifrice or Crest Vivid White Night – HMP containing dentifrice), immersed in saliva overnight, and then brushed in the morning with a CCP (300 gm/50strokes) in a V8 brushing machine after which color was re-measured. In a separate experiment the mPGM plaque mineralization biofilm model was used to assess HMP dentifrice efficacy as compared to CCP dentifrice with application frequencies of 1x/d and alternate days respectively. Anticalculus activity with decreased treatment frequency provides an assessment of retained actions for calculus control. **Results:** Stained pellicle L* CIELAB significantly changed by 1.36(0.36)-CCP and 2.46(0.62)-CVWN showing that HMP dentifrice treatment loosened stains for removal (183 % increase) 8 hours after treatments. mPGM calculus % inhibition showed: CCP control (0); Crest Tartar Control – pyrophosphate 29.8; CVWN ½ treat – 54.0; CVW (standard Vivid White) treatment – 69.5 (all mPGM different significantly @ $p < 0.05$). **Conclusion:** HMP is a strongly retained ingredient, showing stain loosening actions (with overnight wash out) and highly efficient inhibition of calculus mineralization *in vitro*. This supports clinical actions of CVW and CVWN HMP containing dentifrices.

INTRODUCTION

Calcium phosphate surface active builders include molecules which show calcium binding properties in both solution and on intraoral mineral surfaces. CPSAB's such as pyrophosphate and hexametaphosphate show clinical efficacy for reduction of dental calculus formation and removal/prevention of extrinsic dental stains. Crest® Vivid White Night dentifrice contains sodium hexametaphosphate in a silica cleaning base formulated with sodium fluoride for caries prevention. These studies were directed toward demonstrating the physical chemical mechanisms responsible for CVWN clinical efficacy for the loosening of dental stains and calculus prevention.

MATERIALS AND METHODS

Treatments:

Crest Cavity Protection CCP (NaF, silica abrasive)
Crest Tartar Control CTC (NaF, silica abrasive, sodium pyrophosphate)
Crest Dual Action Whitening CDAW
(NaF, silica, sodium hexametaphosphate)
Crest Vivid White CVW (NaF, silica, sodium hexametaphosphate)
Crest Vivid White Night CVWN (NaF, silica, sodium hexametaphosphate)

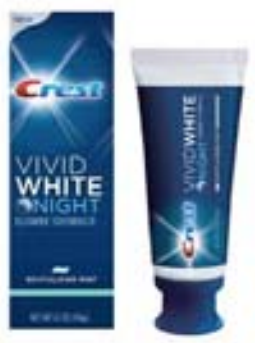

Crest
VIVID
WHITE
NIGHT

MATERIALS AND METHODS

HAP Powder Stain:

Protocol followed model described by Baig et al., J Clin Dent 13: 19-24, 2002. Stain prevention: BioRad HAP chromatography grade pre treated with supernatants of dentifrice/water slurries, centrifuged, washed and then post treated with concentrated tea solution.

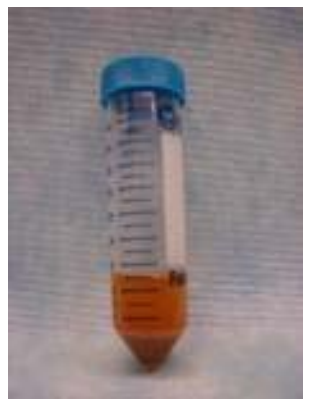

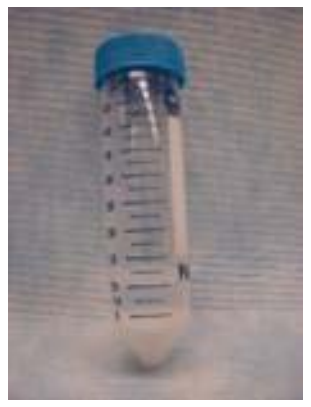

Stain removal: HAP pre treated with concentrated tea solution then post treated with supernatants of dentifrice/water slurries, centrifuged, washed. Powder color compared visually and by digital color analysis (Fuji HC1000 CCD high resolution digital camera with standard lighting)

mPGM Plaque Calcification Tartar Control Assessment:

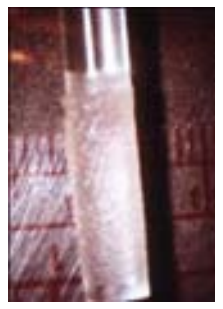

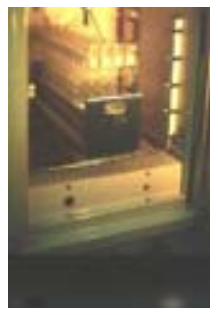

Evaluation for anticalculus activity followed the modified Plaque Growth and Mineralization methodology published previously J Clin Dent 13: 33-37, 2002. Dentifrice slurry treatments of saliva developed plaque biofilm on glass slides is cycled with saliva/artifical saliva incubation and plaque calciuim acquisition is determined following 10 days by atomic absorbance.

Stain Loosening *In Vitro:*

Stained bovine enamel chips (PCR) were prepared as described by Stookey – J Dent Res 61: 1236-1239, 1982. Stain/tooth color was measured with a Fuji X-2000 in CIELAB coordinates and specimens were stratified with respect to initial L* color.

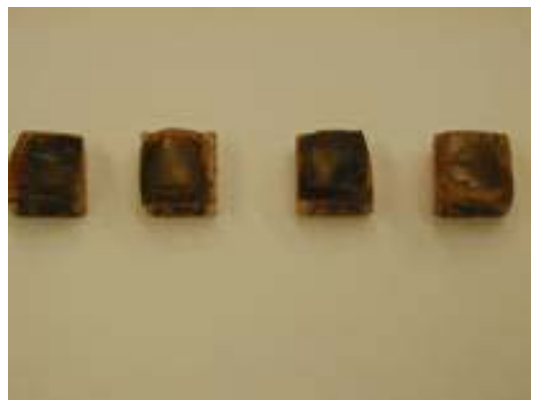

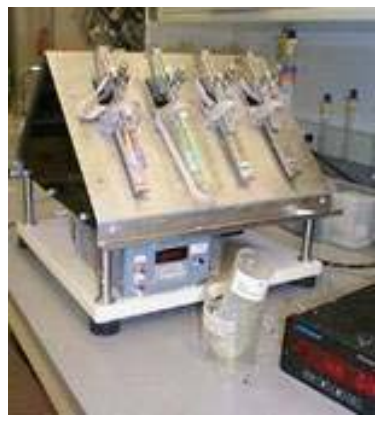

Stained specimens were cycled in a treatment regimen including: Saliva overnight soak; morning soak in dentifrice/water slurries, 8 hour daily soak in saliva (to simulate overnight period); p.m. brushing with Crest Regular dentifrice slurry (V8 brushing, 300 gm force). 5 days cycling was followed with repeated measurements for tooth stain color.

RESULTS

HAP Powder Stain Prevention Results:

Powder Stain Acquistion Following Topical Treatments

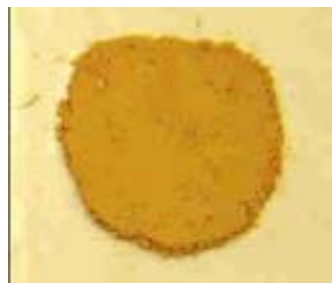

Crest Cavity Protection

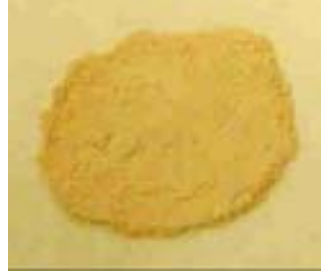

Crest Tartar Control

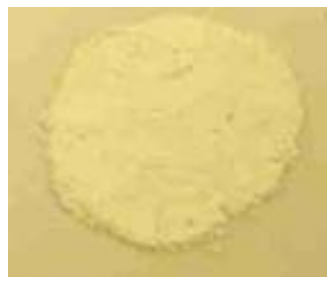

Crest Vivid White Night

mPGM Plaque Calcification Tartar Control Assessment:

Effects of Treatments on Plaque Calcification Associated with Tartar Control Efficacy

Treatment	Mean µg Calcium/mg Plaque ± (S.D.)*	% Reduction
Crest Regular	918.86 ± (71.60) a	00
Crest Tartar Control	645.00 ± (71.10) b	29.8
Crest Vivid White Night - Treated 1/2 standard	423.27 ± (25.00) c	54.0
Crest Dual Action Whitening	302.93 ± (12.93) d	67.0
Crest Vivid White	280.22 ± (15.91) e	69.5

*Means ± (standard deviations), n=4
**Means with different letter designation are significantly different ($p < 0.05$) by the Tukey HSD test.

Stain Loosening *In Vitro*:

Effects of CVWN on Stain Removal - Overnight Protocol Standard PCR Stain

Treatment	ΔL* (Std. Dev)	% Increase
Crest Regular followed by Crest Regular Brushing in a.m.	1.36 (0.36)	----
Crest Vivid White Night followed by Crest Regular brushing in a.m.	2.46 (0.62)	183

p = 0.0016 (two sample t test two sided comparison)
p = 0.0023 (Wilcoxan rank sum test exact p value)

CONCLUSION

HMP shows stain loosening actions on PCR dental stain simulating overnight clinical actions. CVWN demonstrated antitartar mPGM efficacy with simulation of 1x/day dosing. HAP powder treatments confirm HMP surface chemical anti-stain actions.

MARCH 8-11, 2006

P&G

Research presented at the 84th General Session of the AADR, March 8-11, 2006